IMPRIME PGG IMPROVES THE EFFICACY OF CARBOPLATIN, PACLITAXEL AND CETUXIMAB CHEMOIMMUNOTHERAPY OF ADVANCED NON-SMALL CELL LUNG CANCER (NSCLC)

Michael Thomas MD[1], Parvis Sadjadian MD[2], Jens Kollmeier MD[3], Zhonglin Hao MD[4], Nandita Bose PhD[5], Mary Antonysamy PhD[5], Myra Patchen PhD[5], Jamie Lowe[5], Michelle Gargano[5], Richard D. Huhn MD[5], Keith Gorden[5], Paulette Mattson[5], Folker Schneller MD[6] • Biothera, Inc., Eagan MN USA 55121

ABSTRACT

Imprime PGG is a yeast-derived ß-1,3/1,6 glucan that primes innate immune cells to kill monoclonal antibody (mAb)-targeted cancer cells via a mechanism dependent on complement receptor 3 (CR3). In humans, naturally occurring anti-ß glucan antibodies are required for binding of Imprime PGG to CR3 on immune cells. A quantitative assay has been developed to measure these antibodies in serum; subjects with levels conducive to binding are considered "biomarker positive" (BM^+) and others "biomarker negative" (BM^-). In a Phase 2 study, stage IIIb or IV NSCLC subjects received cetuximab (250 mg/m^2 following initial 400 mg/m^2 loading dose) without (Control, N=30) or with Imprime PGG 4 mg/kg (Imprime, N=60) on Days 1, 8 and 15 of each 3-week treatment cycle; all subjects also received carboplatin (AUC 6) plus paclitaxel (200 mg/m^2) on Day 2 of each cycle for the first 4 to 6 cycles. Maintenance treatment with cetuximab alone or with Imprime was continued in subjects achieving radiographic stable disease or tumor responses (RECIST 1.0). In the efficacy population comprised of all treated subjects who had evaluable baseline and post-baseline scans, median overall survival was 11.2 mo in the control group (N=26), 10.2 mo in the entire Imprime group (N=46) (HR 1.06, p=0.85 vs. control), 16.5 mo in the BM^+ Imprime group (N=15) (HR 0.63, p=0.26 vs. control) and 9.1 mo in the BM^- Imprime group (N=31) (HR 1.35, p=0.35 vs. control). Three-year survival was 0% in the control group, 7% in the entire Imprime group, 17% in the BM^+ Imprime group and 0% in the BM^- Imprime group. The objective response rate (ORR, all partial responses) was 23% in the control group, 48% in the entire Imprime group (p= 0.048 vs. control), 67% in the BM^+ Imprime group (p=0.009 vs. control) and 39% in the BM^- Imprime group (p=0.26 vs. control). Among subjects with squamous cell histology, 6 of 6 BM^+ Imprime subjects had responses compared with 3 of 10 control subjects (p=0.01). Grade 3/4 adverse events occurred in 25 of 29 control subjects (86%) and 46 of 59 Imprime subjects (78%). All adverse events were consistent with toxicities attributable to the cytotoxic drugs or cetuximab. In summary, the addition of Imprime PGG to chemoimmunotherapy with carboplatin, paclitaxel and cetuximab resulted in improved outcomes in BM^+ subjects with respect to increased ORR and extended survival compared to control subjects and had a good safety profile.

INTRODUCTION

Lung cancer is the leading cause of cancer mortality, responsible for an estimated 159,480 deaths in the United States in the year 2013.[1] Non-small cell lung cancer (NSCLC) is a heterogeneous group of various histological subtypes of carcinoma, of which adenocarcinoma and squamous cell carcinoma represent the majority.[2] The prognosis for patients with advanced NSCLC is very poor regardless of chemotherapy. Median overall survival in clinical trials of platinum-based chemotherapy without or with cetuximab for unselected patients with advanced NSCLC ranged from 10 –12 mos. in recent reports.[4-7]

Imprime PGG® (Imprime PGG) is a proprietary soluble yeast-derived ß-glucan that binds and activates the complement receptor 3 (CR3) on innate immune cells (neutrophils and monocytes), directing these cells to kill tumor cells that have been opsonized by iC3b following targeting by anti-tumor monoclonal antibodies (mAb).[8] Numerous studies in human xenograft and syngeneic tumor models in mice have demonstrated that administration of Imprime PGG in combination with anti-tumor mAb treatment reduces tumor growth and extends long-term survival beyond that observed with either agent alone.[9-12] Tumor model studies in various knock-out mouse strains have shown an absolute requirement for CR3 on innate immune cells, the presence of functional complement, and the presence of neutrophils for Imprime PGG mediated antitumor activity.

Binding of Imprime PGG to neutrophils and monocytes is time-, temperature- and serum-dependent.[13] In humans, naturally occurring anti-ß-glucan antibodies (ABA) are required for binding of Imprime PGG to these innate immune cells.[14] Biothera has discovered that that there are minimum ABA levels necessary for this binding. An emzyme-linked immunosorbent assay (ELISA) for measuring serum ABA has been developed to quantitatively identify subjects with ABA levels conducive for Imprime PGG binding to innate immune cells ("biomarker positive") *vs* others ("biomarker negative").

Although not approved by FDA for treatment of NSCLC, cetuximab is an important component of chemotherapy recommendations for NSCLC by the National Comprehensive Cancer Network.[15] BT-LCA-0822 study was a phase 2 randomized clinical trial in which subjects with advanced NSCLC were enrolled and treated with carboplatin, paclitaxel and cetuximab either without or with Imprime PGG with data analyzed overall as well as according to Imprime PGG biomarker status and histology status.

BIOTHERA
the immune health company

AFFILIATIONS

[1]Thoraxklinik im Universitätsklinikum Heidelberg, Heidelberg, DE; [2]Johannes Wesling Klinikum Minden, Minden, DE; [3]Helios Clinic Emil von Behring, Berlin, DE; [4]Georgia Health Sciences University, Augusta, GA, USA; [5]Biothera, Inc., Eagan MN USA 55121; [6]Klinikum rechts der Isar of Technical University Munich, Munich, DE.

REFERENCES

[1]www.cancer.org; [2]www.cancer.gov; [3]Pirker R et al., *The Lancet* 373:1525, 2009; [4]Lynch TJ et al., *J. Clin. Oncol.* 28:911, 2010; [5]Butts CA et al., *J. Clin. Oncol.* 25:5777, 2007; [6]Pirker R et al., *The Lancet Oncology* 13:33, 2012; [7]Hirsch FR et al., *J. Clin. Oncol.* 21:3798, 2003; [8]Li B et al., *J Immunol* 177:1661, 2006; [9]Liu J, Gunn L, Hansen R & Yan J, *Exp. Mol. Pathol.* 86:208, 2009; [10]Li B et al., *Cancer Res* 67:7421, 2007; [11]Salvador C et al., *Clin Cancer Res.* 14:1239, 2008; [12]Qi C et al., *Blood* 117:6825, 2011; [13]Bose N et al., *Front. Immunol.* 4:230, 2013; [14]Bose N et al., Cancer Immunotherapy Conference, Keystone Symposium, British Columbia, Canada, March 2013; [15]www.nccn.org

METHODS

STUDY DESIGN:

90 subjects with previously untreated stage IIIb or IV NSCLC were enrolled and randomized to receive cetuximab (250 mg/m^2) following initial 400 mg/m^2 loading dose) without (Control, N=30) or with Imprime PGG 4mg/kg (Imprime PGG, N=60) on Days 1, 8 and 15 of each 3-week treatment cycle; all subjects also received carboplatin (AUC 6) plus paclitaxel (200 mg/m^2) on Day 2 of each cycle for the first 4 to 6 cycles. Following the completion of at least the initial 4 treatment cycles (but no more than 6), subjects who experienced stable disease, or a complete or partial response, were eligible to continue on maintenance treatment with Imprime PGG and cetuximab in the Imprime PGG Arm or cetuximab alone in the Control Arm.

Objective response rate (ORR) was the primary endpoint. Overall survival (OS), as well as safety/tolerability and pharmacokinetics were secondary endpoints. Data from Imprime PGG subjects were evaluated overall as well as by biomarker status. A sub-analyses according to histology (squamous vs non-squamous) was also performed.

TUMOR MEASUREMENTS:

Tumor measurements and determination of tumor response/progression were performed according to modified RECIST v1.0. The Primary Efficacy Population (PEP) consisted of subjects who had an evaluable baseline CT scan and at least one evaluable post-baseline scan (unless discontinued earlier due to progressive disease).

IMPRIME PGG BIOMARKER ANALYSIS:

Baseline sera from subjects in the Imprime PGG Arm were analyzed for levels of naturally occurring ABA by a enzyme-linked immunosorbent assay (ELISA) in which a serum standard with a predetermined ABA level was concurrently assayed. Optical density measurements of the subjects' sera were converted to values of antibody concentration corresponding to the serum standard at each dilution level and values were averaged across dilutions to yield a Relative Antibody Unit (RAU) value for each serum sample. Subjects were scored as biomarker positive if their RAU level was greater than or equal to threshold levels previously determined to predict substantial Imprime PGG binding to neutrophils and monocytes in a study in healthy volunteers; the remaining subjects were considered as biomarker negative.

Key inclusion and exclusion criteria are shown in Table1.

Inclusion	Exclusion
• histologically or cytologically confirmed stage IIIB (malignant pericardial or pleural effusion) or stage IV non-small cell lung cancer • measurable disease, defined as at least one tumor that fulfills the criteria for a target lesion according to RECIST • age between 18 and 75 y.o., inclusive • ECOG performance status of 0 or 1 • life expectancy >3 mo. • adequate hematologic, renal and hepatic function (see protocol for specifics)	• prior treatment with chemotherapy for lung cancer • previous radiation >30% active bone marrow or any radiation therapy within 3 weeks of Day 1 • known hypersensitivity to Bakers yeast, or active yeast infection • previous exposure to Betafectin or Imprime PGG • active infection • medical diagnoses/conditions at the time of screening: • CNS metastases • peripheral neuropathy ≥grade 2 (any cause) • fever >38.5° C within 3 days prior to screening or Day 1, initial dosing • known HIV/AIDs, hepatitis B, hepatitis C, connective tissue or autoimmune disease, or other clinical diagnosis, ongoing or intercurrent illness that in the physician's opinion could interfere with participation • history of any of the following medical diagnoses/conditions: • myocardial infarction or unstable angina or uncontrolled disease or condition related to or impacting cardiac function (e.g., unstable angina, congestive heart failure) within the previous 6 months • second malignancy within the previous 5 years, other than basal cell carcinoma, cervical intraepithelial neoplasia or curatively treated prostate cancer with PSA <2.0 ng/mL • known hypersensitivity to cetuximab, murine proteins or any component of cetuximab • concurrent investigational therapy or investigational therapy within 30 days prior to the first scheduled dosing day • previous organ or progenitor/stem cell transplant

Table1.

RESULTS

SAFETY:

Of the 90 randomized subjects, one in each arm failed to receive any protocol therapy, leaving 29 subjects in the Control Arm and 59 subjects in the Imprime PGG Arm for safety assessment. Adverse events were consistent with toxicities known to be attributable to the backbone chemotherapy of carboplatin, paclitaxel and cetuximab.

The overall frequency of Grade 3 or 4 adverse events in the Safety Population is shown in Table 2.

	Control (N=29)	All Imprime PGG (N=59)
Subjects with Grade 3 or 4 AEs	25 (86%)	46 (78%)
Deaths due to AEs	0 (0%)	2* (3%)

Table 2. *One subject with neutropenic sepsis and acute renal failure considered unlikely related to Imprime PGG and one subject with a pleural effusion considered unlikely related to Imprime PGG.

The frequencies of the most common Grade 3 or 4 adverse events (≥5% in either treatment arm) are shown in Table 3.

	Control (N=29)	All Imprime PGG (N=59)
Blood and Lymphatic System Disorders		
Neutropenia	14 (48%)	19 (32%)
Leukopenia	9 (31%)	9 (15%)
Anemia	2 (7%)	3 (5%)
Thrombocytopenia	2 (7%)	2 (3%)
Gastrointestinal Disorders		
Diarrhea	3 (10%)	0 (0%)
General Disorders and Administration Site Conditions		
Fatigue	0 (0%)	4 (7%)
Investigations		
Hemoglobin Decreased	0 (0%)	4 (7%)
Blood Phosphorus Decreased	2 (7%)	1 (2%)
White Blood Cell Count Decreased	2 (7%)	0 (0%)
Metabolism and Nutrition Disorders		
Hypokalemia	1 (3%)	3 (5%)
Nervous System Disorders		
Polyneuropathy	2 (7%)	4 (7%)
Respiratory, Thoracic and Mediastinal Disorders		
Pulmonary Embolism	2 (7%)	4 (7%)
Pleural Effusion	0 (0%)	3 (5%)
Skin and Subcutaneous Tissue Disorders		
Rash	3 (10%)	2 (3%)

Table 3.

Demographic features of the Primary Efficacy Population are summarized in Table 4.

	Control (N=26)	All Imprime PGG (N=46)	Biomarker Positive Imprime PGG (N=15)	Biomarker Negative Imprime PGG (N=31)
Age (yrs.)	61.8±7.2	59.9±10.1	60.3±12.5	59.6±8.9
Gender				
Male	16 (62%)	34 (74%)	12 (80%)	22 (71%)
Female	10 (38%)	12 (26%)	3 (20%)	9 (29%)
Ethnic origin				
White	26 (100%)	44 (96%)	14 (93%)	30 (97%)
Black	0 (0%)	1 (2%)	0 (0%)	1 (3%)
Other	0 (0%)	1 (2%)	1 (7%)	0 (0%)
ECOG Performance Status				
0	10 (38%)	17 (37%)	6 (40%)	11 (35%)
1	15 (58%)	29 (63%)	9 (60%)	20 (65%)
missing	1 (4%)	0 (0%)	0 (0%)	0 (0%)

Table 4.

Tumor response rates for the Primary Efficacy Population, as sub-divided by tumor histology, are shown in Table 5. *p*-values shown are vs. control.

Objective Response Rate	Control	All Imprime PGG	Biomarker Positive Imprime PGG	Biomarker Negative Imprime PGG
All histologies	23% (6/26)	48% (22/46) p=0.047	67% (10/15) p=0.009	39% (12/31) p=0.259
Squamous	30% (3/10)	62% (8/13) p=0.214	100% (6/6) p=0.011	29% (2/7) p=1.00
Non-Squamous	19% (3/16)	42% (14/33) p=0.123	44% (4/9) p=0.205	42% (10/24) p=0.177

Table 5.

Median overall survivals (mOS) for the Primary Efficacy Population, as divided by tumor histology, are shown in Table 6. Hazard ratios and *p*-values shown are vs. control.

mOS (mos.)	Control	All Imprime PGG	Biomarker Positive Imprime PGG	Biomarker Negative Imprime PGG
All histologies	11.2 (N=26)	10.2 (N=46) HR 1.06 p=0.853	16.5 (N=15) HR 0.63 p=0.262	9.1 (N=31) HR 1.35 p=0.345
Squamous	12.2 (N=10)	13.6 (N=13) HR 1.01 p=0.983	15.6 (N=6) HR 0.62 p=0.469	5.2 (N=7) HR 1.40 p=0.568
Non-Squamous	9.8 (N=16)	9.6 (N=33) HR 0.99 p=0.984	18.8 (N=9) HR 0.56 p=0.316	9.3 (N=24) HR 1.21 p=0.628

Table 6.

The proportion of patients surviving over time in the Safety population is shown in Table 7.

Survival	Control (N=29)	All Imprime PGG (N=59)	Biomarker Positive Imprime PGG (N=22)	Biomarker Negative Imprime PGG (N=37)
1 yr.	47%	49%	65%	36%
2 yr.	14%	18%	26%	14%
3 yr.	0%	7%	17%	0%

Table 7.

The graphical display of the Kaplan Meyer analysis comparing the survival of the biomarker Positve Imprime PGG Arm to the Control Arm in the Primary Efficacy Population is shown in Figure 1.

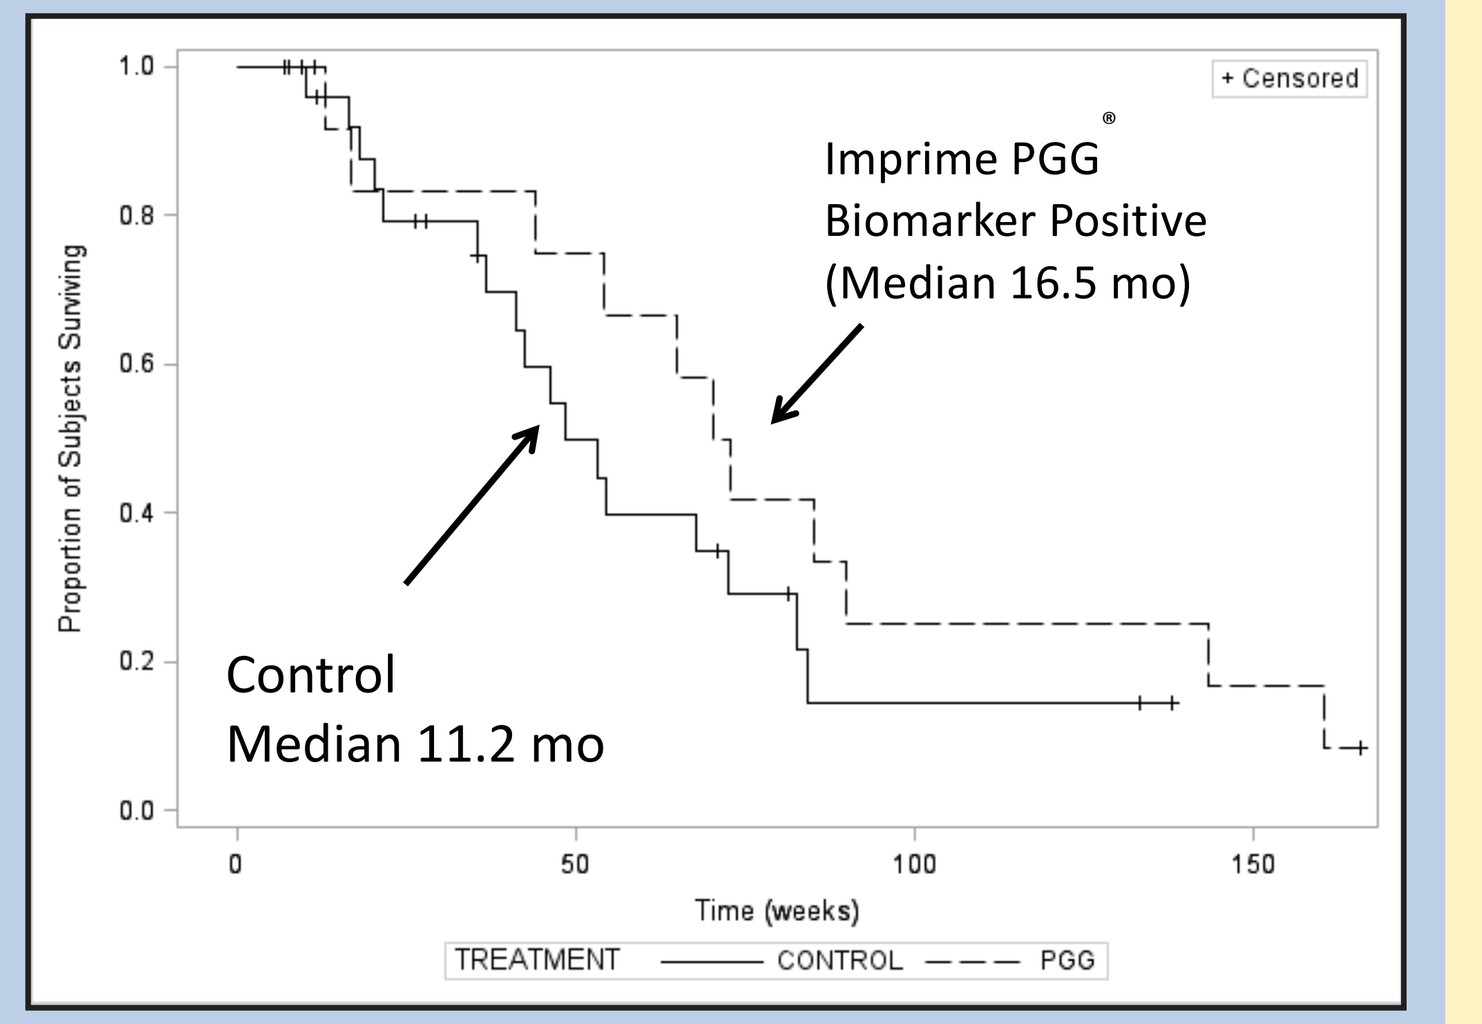
+ Censored
Imprime PGG®
Biomarker Positive
(Median 16.5 mo)
Control
Median 11.2 mo
Proportion of Subjects Surviving
1.0
0.8
0.6
0.4
0.2
0.0
0
50
100
150
Time (weeks)
TREATMENT
CONTROL
PGG

Figure 1.

SUMMARY AND COMMENTS

- The addition of Imprime PGG® to the backbone therapy of cetuximab plus carboplatin and paclitaxel as first line therapy for Stage IIIB/IV NSCLC resulted in improved outcomes in the Imprime PGG® biomarker positive subjects compared to the control subjects:
 - Objective response rates (the primary endpoint of the study) were significantly increased overall as well as in the squamous histology subpopulation.
 - Although the study was not powered for survival:
 - Survival was improved approximately 5 months overall.
 - Improved survival was also observed in both squamous and non-squamous histology subpopulations.
 - Survival hazard ratios ranged from 0.5 - 0.6, representing a strong positive trend for improvement.
 - Improved outcomes seen in the Imprime PGG® biomarker positive subjects were not biased by prognostic demographic factors.
- Imprime PGG® exhibited a good safety profile – common adverse events were generally characteristic of those of cetuximab plus carboplatin and paclitaxel.
- Further work to ascertain the predictive significance of the biomarker is being planned.
- Additional trials to confirm the benefit of Imprime PGG combined with anti-tumor mAbs for treatment of advanced cancer are ongoing and being planned.