A Multicenter, Open-label, Phase II Study of Imprime PGG (PGG Beta-Glucan) and Pembrolizumab in Patients With Advanced Melanoma Failing Front-line Treatment with Checkpoint Inhibitors or Triple Negative Breast Cancer Failing Front-line Chemotherapy for Metastatic Disease

Authors: J. Iglesias[1], R. Prathikanti[2], B. Ma[2], P. Mattson[2], D. Kedrowski[2], J. Lowe[2], N. Bose[2], K. Ertelt[2], N. Ottoson[2], M. Uhlik[2], J. Graff[2], R. Huhn[2], M. Chisamore[3]

Affiliations: [1]Apobiologix, Toronto, Canada; [2]Biothera Pharmaceuticals, Inc., Eagan, MN, USA; [3]Merck & Co., Inc., Kenilworth, NJ, USA

Background

There are few attractive options for treatment of checkpoint inhibitor (CPI)-refractory or -relapsed **metastatic melanoma (MEL).** Cytotoxic chemotherapy is ineffective. In a recent study, nivolumab combined with ipilimumab provided a response rate of 57.6 % and mPFS > 11 mos. in previously untreated patients (pts); however, this combination resulted in high rates of severe adverse effects including immune-related adverse effects.[1]

The only treatment options for advanced **triple-negative (ER/PR-, HER2-) breast cancer (TNBC)** are cytotoxic chemotherapy regimens which are poorly effective and highly toxic. CPIs, including pembrolizumab, have been studied in pts with advanced TNBC with modest effectiveness. In the KEYNOTE-012 trial, the overall response rate in 27 evaluable pts was 18.5%.[2]

Less toxic approaches to therapy of CPI-relapsed or –refractory advanced MEL are needed as are improved treatment options for refractory and relapsed TNBC.

Rationale

Imprime PGG is a yeast-derived soluble beta-glucan that acts as a pathogen-associated molecular pattern to stimulate innate immune cells and coordinates a multi-faceted immune response in the tumor microenvironment[3]
- Generates immunomodulatory cytokines and chemokines
- Repolarizes M2 to M1 phenotype of macrophages
- Enhances dendritic cell activation

Imprime PGG:
- Effectively pairs with anti-PD-1 and -PD-L1 antibodies to induce immunity against syngeneic tumors in mouse models[4]
- Is complexed by naturally occurring IgG anti-beta glucan antibodies (ABA) which are required for its activity; serum ABA level is a potential predictive biomarker for Imprime PGG activity[5,6]
- Has been well-tolerated in clinical trials in healthy volunteers and cancer pts[7,8]

Pembrolizumab (KEYTRUDA; [Pembro]), is a humanized mAb against programmed death receptor-1(PD-1) approved for advanced MEL, advanced NSCLC, recurrent or metastatic head and neck cancer, and classical Hodgkin Lymphoma.

IMPRIME-1 is a multicenter, open-label, Phase 2 clinical trial of Imprime PGG (PGG beta-glucan) in combination with pembrolizumab in pts with advanced MEL failing front-line treatment with CPIs or TNBC failing front-line chemotherapy for metastatic disease.

This trial is registered with Clinicaltrials.gov: NCT02981303 and is enrolling investigative sites located in the United States.

Imprime PGG Impacts Multiple Points of the Cancer Immunity Cycle and Drives Enhanced Responsiveness to CPI

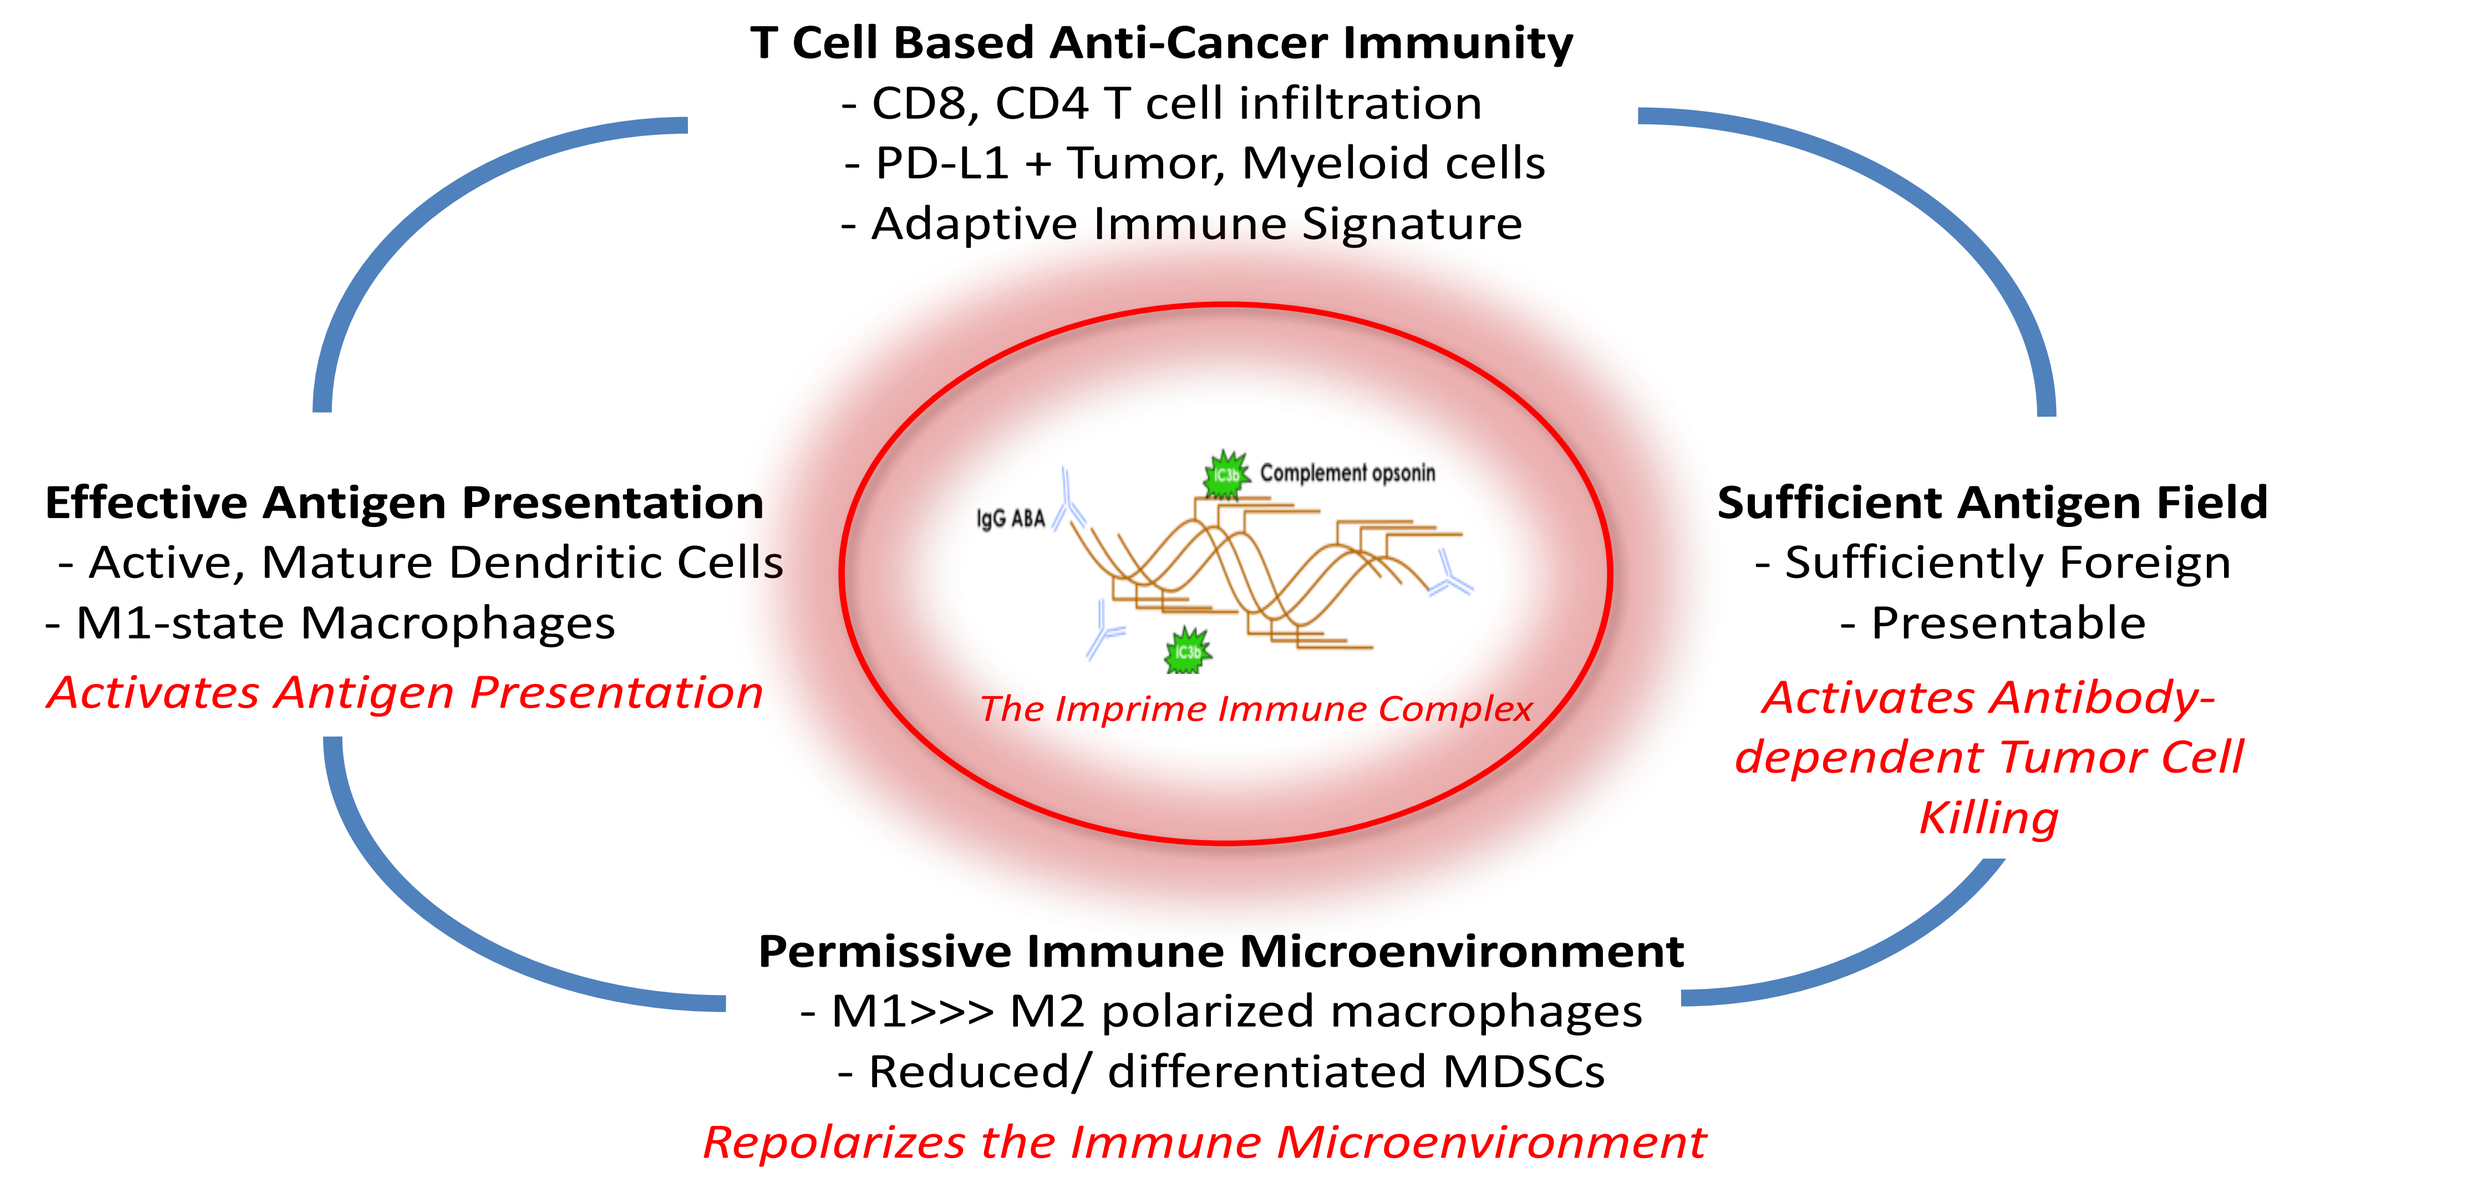

T Cell Based Anti-Cancer Immunity
- CD8, CD4 T cell infiltration
- PD-L1 + Tumor, Myeloid cells
- Adaptive Immune Signature
Effective Antigen Presentation
- Active, Mature Dendritic Cells
- M1-state Macrophages
Activates Antigen Presentation
IgG ABA
Complement opsonin
The Imprime Immune Complex
Sufficient Antigen Field
- Sufficiently Foreign
- Presentable
Activates Antibody-dependent Tumor Cell Killing
Permissive Immune Microenvironment
- M1>>> M2 polarized macrophages
- Reduced/ differentiated MDSCs
Repolarizes the Immune Microenvironment

Hypothesis

Imprime PGG in combination with pembrolizumab will restore (for MEL) or enhance (for TNBC) sensitivity to checkpoint inhibitors (CPI) by appropriate and effective stimulation of the pt's innate and adaptive immune systems in those pts who have failed first-line therapy

Study Design

Seventy-one (71) pts will be enrolled into the study, 29 pts with advanced MEL and 42 pts with TNBC.

The study design is based on hypothesis testing of the ORR for pembrolizumab + Imprime PGG
- ***MEL:*** null hypothesis of ORR ≤ 5% vs. alternative hypothesis of ORR ≥ 23%
- ***TNBC:*** null hypothesis of ORR ≤ 19% vs. alternative hypothesis of ORR ≥ 40%

The study incorporates Simon's optimal 2-stage design with sample size fixed at 12 pts for each indication in Stage 1, while Stage 2 will enroll an additional 17 MEL pts and 30 TNBC pts
- Type I error of 0.05 and 90% power for both MEL and TNBC
- At Stage 1, in the event there are a total of > 4 (or > 33%) of pts with Grade 3/4 adverse events in Cycle 1, Stage 1 will be repeated at a reduced dose of 2 mg/kg with an additional cohort of 12 pts in each arm
- Criterion for proceeding to Stage 2:
 - ≤ 4 (or ≤ 33%) pts with Grade 3/4 adverse events in Cycle 1 within either tumor type
 - At least 1 response in MEL pts and 2 responses in TNBC pts
 - Either arm may proceed independently
- Rejection of the null hypothesis will require documenting at least 4 objective responses in MEL and 13 objective responses in TNBC at the conclusion of Stage 2

Study Design Cont.

Study Schema

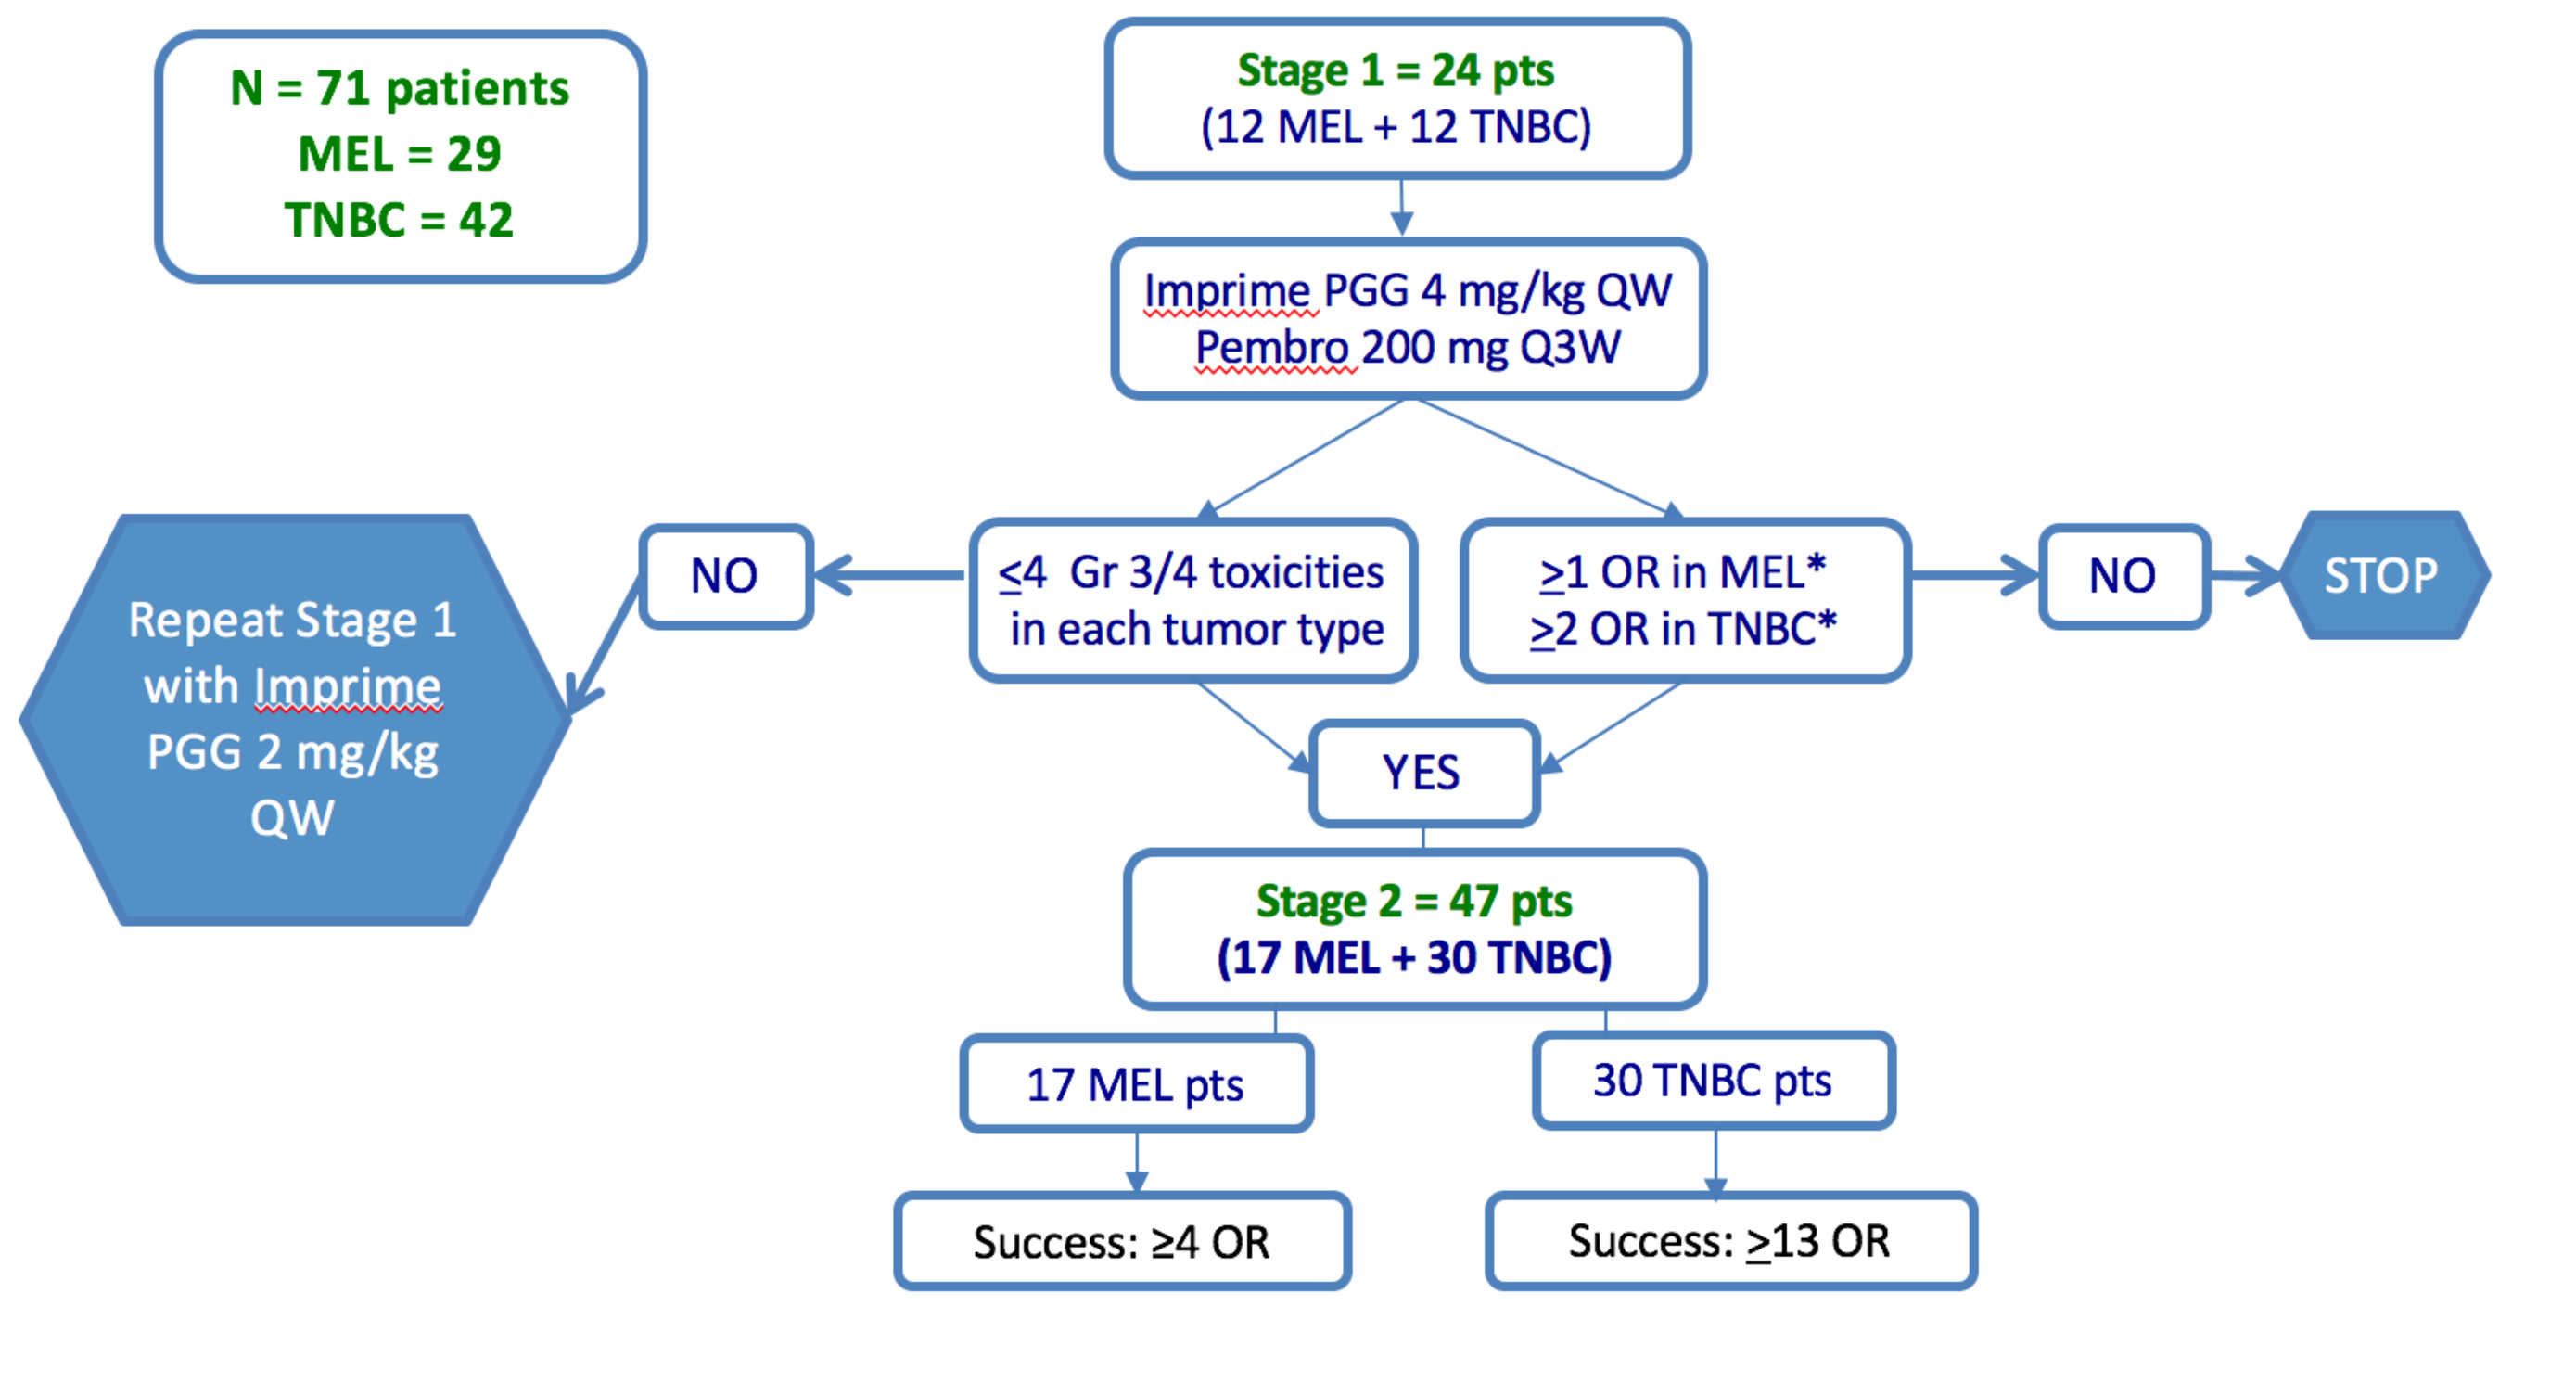

N = 71 patients
MEL = 29
TNBC = 42
Stage 1 = 24 pts
(12 MEL + 12 TNBC)
Imprime PGG 4 mg/kg QW
Pembro 200 mg Q3W
Repeat Stage 1 with Imprime PGG 2 mg/kg QW
NO
≤4 Gr 3/4 toxicities in each tumor type
≥1 OR in MEL*
≥2 OR in TNBC*
NO
STOP
YES
Stage 2 = 47 pts
(17 MEL + 30 TNBC)
17 MEL pts
30 TNBC pts
Success: ≥4 OR
Success: ≥13 OR

*Arms may proceed independently into Stage 2. OR, overall response

Study Treatment:
- Imprime PGG 4 mg/kg IV is administered weekly
- Pembrolizumab 200 mg IV is administered on Day 1 of each 3-week cycle
- Pts will receive treatment until disease progression per RECIST 1.1 or intolerable toxicity

Study Assessments:
- On-study assessments
 - Tumor imaging/response: CT of chest, abdomen and pelvis Q6wks
 - RECIST v1.1 for tumor response
 - irRECIST for clinical management of pts
 - Physical exam, vital signs, ECOG performance status, clinical safety and laboratory assessments conducted throughout
- Post-treatment assessments
 - Monitoring every 2-months for survival until death or until study completion
 - Pts who discontinued treatment without evidence of PD will have CT scans Q6wks until PD is noted
- Tumor biopsies will be collected
 - at baseline (diagnostic pathology report +/- archival tumor biopsy sample)
 - on-study before first dose, between Cycles 2 and 3, and at the time of response or if no response, at the End of Study Treatment Visit
- Sampling will be done throughout study to support translational endpoints
- Sparse PK sampling will be assessed on Cycle 1 Day 1 and Cycle 3 Day 1

Key Study Endpoints

Primary Efficacy Endpoint
- Overall Response Rate (ORR) in pts with advanced MEL or metastatic TNBC

Secondary Efficacy Endpoints
- Time to response (TTR)
- Complete response rate (CRR)
- Duration of overall response (DoR)
- Progression-Free Survival (PFS) and PFS rate at 6 months and 1 year
- Overall survival (OS) and OS rate at 1 year

Safety Endpoints
- Incidence of treatment-emergent adverse events and laboratory abnormalities

Pharmacokinetic Endpoints
- Profile pharmacokinetic (PK) data

Exploratory Immune Correlative Objectives
- Determine ORR and PFS based on irRECIST
- Correlate the following with OR and treatment outcome:
 - Levels of baseline serum anti-β-glucan antibody (ABA)
 - Changes in immune cell activation markers in tumor biopsy samples and peripheral blood immune cells
 - Changes in the tumor immune microenvironment including tumor-infiltrating lymphocytes and tumor-infiltrating myeloid cells
 - PD-L1 expression in tumor biopsy samples (in tumor and myeloid cells)

Key Eligibility Criteria

Adults who have:
- ***MEL:*** histologically or cytologically confirmed diagnosis of unresectable Stage III or metastatic (Stage IV) MEL not amenable to local therapy and irrespective of PD-L1 status; or
- ***TNBC:*** histologically or cytologically confirmed diagnosis of metastatic (Stage IV) TNBC irrespective of PD-L1 status
 - TNBC is defined as negative IHC ER/PR assays and HER2 negative (IHC 0 or 1+, or 2+ by IHC confirmed negative by FISH)

Key Eligibility Criteria Cont.

- Documented objective radiographic or clinical disease progression after:
 - ***MEL:*** PD-1/PD-L1 +/- anti-CTLA-4 inhibitor therapy
 - ***TNBC:*** 1 or more prior lines of chemotherapy for metastatic disease*
- At least one radiologically measurable lesion per RECIST v1.1 imaged by CT scan or MRI within 28 days of study treatment
- Screening peripheral blood levels of IgG anti-β-glucan antibody (ABA) of ≥ 20 mcg/mL
- Willing to consider providing fresh tissue for biomarker analysis and/or assessment

Excluded:
- Pts who had a prior anti-cancer monoclonal antibody (except immune CPI in the case of MEL pts) within 30 days prior to start of study treatment, or failed to recover to CTCAE Grade 1 or better from adverse events related to prior therapies
- TNBC pts who received prior therapy with an anti-PD-1, anti-PD-L1, anti-CTLA-4, or anti-PD-L2 agent

- Eastern Cooperative Oncology Group (ECOG) performance status of 0 or 1
- Adequate organ function
- Documented informed consent
- Life Expectancy of ≥ 3 months

* Inclusion criteria language pending approval of a protocol amendment; current protocol language reads, "after at least 1 line of chemotherapy for metastatic disease"

General Statistical Methods

Interim Analysis
- At the end of Stage 1 for each tumor type
- Early stopping for futility, not efficacy

Efficacy Analysis
- ORR and CRR: point estimate along with 95% CI
- PFS, OS, DoR and TTR: descriptive summary using Kaplan-Meier method

Safety Analysis
- Summaries of adverse events, hematology and clinical chemistry values, ECOG performance status

Exploratory Analysis
- Correlation between biomarker data with ORR and other clinical endpoints

Study Oversight

A Trial Steering Committee will provide study oversight.

Acknowledgements

We would like to thank all patients, investigators and study staff for their participation in this clinical trial.

Disclosures

This trial is sponsored by Biothera Pharmaceuticals, Inc. Author disclosures:
- J. Iglesias is an employee of Apobiologix
- R. Prathikanti, B. Ma, P. Mattson, D. Kedrowski, J. Lowe, N. Bose, K. Ertelt, N. Ottoson, M. Uhlik, J. Graff, and R. Huhn are sponsor employees holding stock options and/or grants
- M. Chisamore is an employee of Merck & Co. holding stock options and/or grants

Corresponding Author

rhuhn@biothera.com

References

1. Larkin J, Chiarion-Sileni V, Gonzalez R, et al., N Engl J Med 373:23, 2015
2. Nand R, Chow LQM, Dees EC, et al., J Clin Oncol 34:2460, 2016
3. Bose N, Gorden K, Chan A, et al., Cancer Immunol Res 5: 3 Suppl B29, 2017
4. Fraser K, Chan A, Fulton R, et al., Cancer Res 76:14 Suppl 2335, 2016
5. Bykowski Jonas A, Yokoyama Y, Ottoson N, Bose N, Graff J, Cancer Res 76:14 Suppl LB-080, 2016
6. Chan ASH, Bykowski Jonas A, Qui X, et al., PLoS ONE 11(11):e0165909, 2016
7. Bose N, Ottoson N, Harrison B, et al., J Clin Oncol 35:suppl 7S: abstract 33, 2017
8. Thomas M, Sadjadian P, Kollmeier J, et al., Invest New Drugs 35:345, 2017

IMPRIME
Cancer Immunotherapy Clinical Trials

Copies of this poster obtained through Quick Response (QR) code are for personal use and may not be reproduced without permission from ASCO® and the author of this poster. For questions, please contact permissions@asco.org.

Abstract TPS3105

2017 ASCO Annual Meeting, June 2-6, Chicago, IL